#4318

Development of a novel dual agonist Surrobody™ that simultaneously activates both death receptors DR4 and DR5 and induces cancer cell death with high potency

Snezana Milutinovic[1], Sihong Zhou[2], Carina Wimer[1], Paul W. Diaz[1], Arun K. Kashyap[2], Michael Horowitz[2], Ramesh R. Bhatt[2], **John C. Reed[1]**

[1]Sanford-Burnham Medical Research Institute, La Jolla, CA, [2]Sea Lane Biotechnologies, Mountain View, CA

Sanford|Burnham Medical Research Institute

Abstract

Death receptors of the Tumor Necrosis Factor (TNF) family are found on surface of most cancer cells and their activation typically kills cancer cells through the stimulation of the extrinsic apoptotic pathway. The endogenous ligand for death receptors-4 and -5 (DR4 and DR5) is tumor necrosis factor-related apoptosis-inducing ligand, TRAIL (Apo2L). TRAIL is expressed by many cell types, including Natural Killer (NK) cells that are involved in the immune surveillance and elimination of cells in vivo. Given that most normal untransformed cells are not susceptible to TRAIL-induced apoptosis, activators of death receptors have emerged as promising cancer therapeutic agents. One strategy to stimulate death receptors in cancer patients has been to use soluble human recombinant TRAIL protein, but this agent has limitations of a short half-life and decoy receptor sequestration. Another strategy that attempted to evade decoy receptor sequestration and to have greater pharmacokinetic properties was to generate DR4 or DR5 agonist antibodies. The resulting monoclonal agonist antibodies overcame the limitations of short half-life, but exhibited the limitation of activating only one of the two death receptors. The Surrobody™ binding protein is a novel therapeutic structure based upon the pre-B cell receptor capable of binding targets with high affinity and specificity. We exploited a previously discovered Surrobody binding protein that potently activates both DR4 and DR5 in vitro and in vivo. Here we show characterization of a dual agonist Surrobody that offers a powerful and likely more reliable strategy for development of therapeutics based on stimulation of TRAIL receptors.

Background

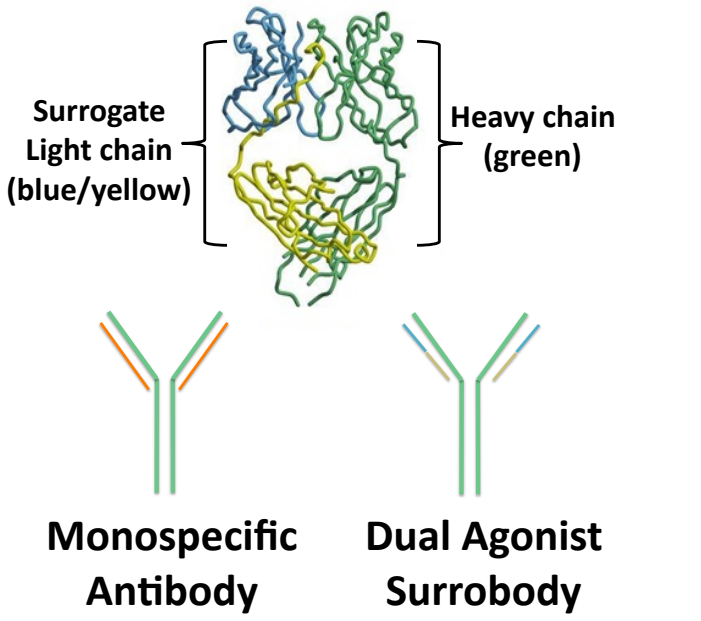
Surrogate Light chain (blue/yellow)
Heavy chain (green)
Monospecific Antibody
Dual Agonist Surrobody

We assessed a collection of more than 300 unique clones found by panning a human phage displayed Surrobody™ library against human DR4 as previously described to identify a DR4 and DR5 crossreactive Surrobody. The crossreactive clone was then reformatted and expressed as a bivalent Surrobody using full length heavy chains in HEK293 cells as previously described (1, 2). The resulting Surrobodies, termed SgGs (for "Surroglobulins"), were transiently expressed at levels routinely yielding approximately 100 mg/L of culture and formed the basis of all subsequent analysis.

Results

Figure 1. Surrobody is a more potent inducer of cancer cell death than anti-DR4 or anti-DR5 monospecific antibodies.

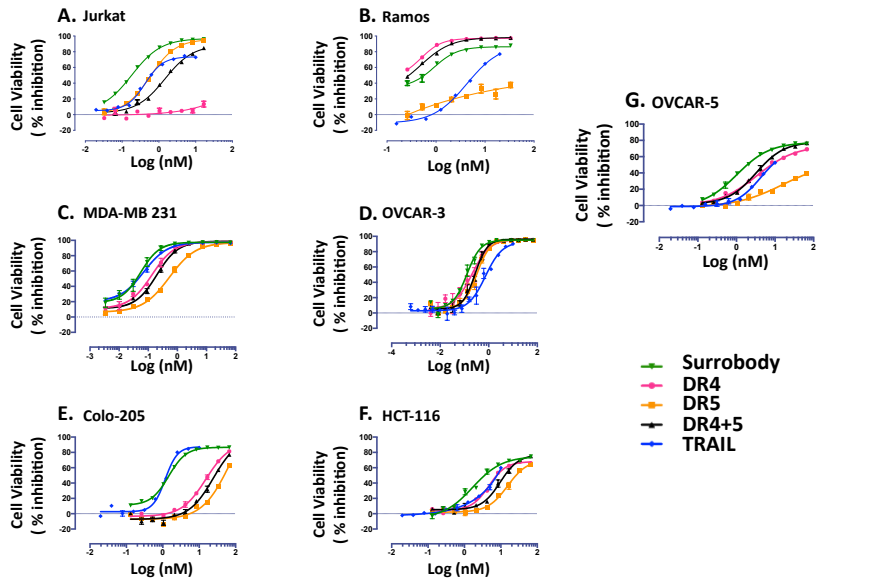
A. Jurkat
B. Ramos
C. MDA-MB 231
D. OVCAR-3
E. Colo-205
F. HCT-116
G. OVCAR-5
Cell Viability (% inhibition)
Log (nM)
Surrobody
DR4
DR5
DR4+5
TRAIL

Various TRAIL-sensitive cancer cell lines were plated at a density of 750 cells/well in 384 wells. The next day cells were cultured with increasing concentrations of monospecific DR4 antibody (pink), monospecific anti-DR5 antibody (orange), the combination of anti-DR4 and anti-DR5 antibodies (black), DR4/DR5 dual agonist Surrobody (bright green), or TRAIL (blue) for 48 hr. Relative cell viability was estimated from cellular ATP measurements using the Cell Titer Glow reagent (Promega), expressing data as % inhibition of cell survival (mean ± SEM; n= 4). Data are shown for Jurkat (A), Ramos (B), MDA-MB-231(C), Ovcar-3 (D), Colo-205 (E) HCT116 (F), Ovcar-5 (G) cell lines.

Figure 2. Smac mimics sensitize TRAIL-resistant tumor cells.

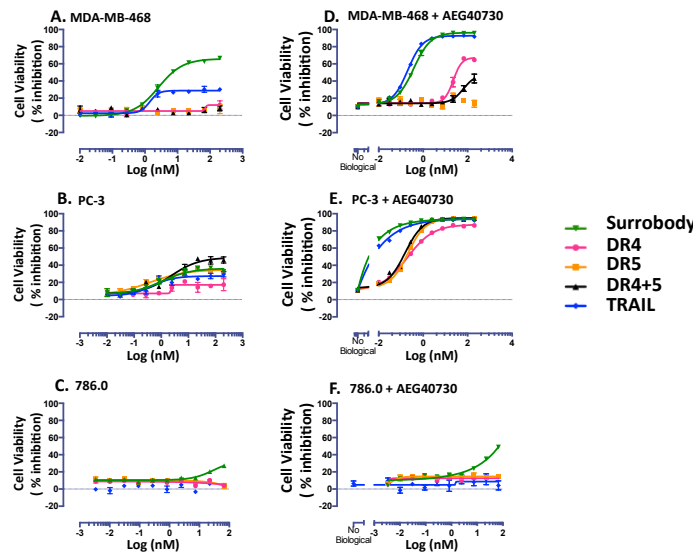
A. MDA-MB-468
B. PC-3
C. 786.0
D. MDA-MB-468 + AEG40730
E. PC-3 + AEG40730
F. 786.0 + AEG40730
Cell Viability (% inhibition)
Log (nM)
No Biological
Surrobody
DR4
DR5
DR4+5
TRAIL

TRAIL resistant cell lines, including MDA-MB-468 (A), PC3 cells (B) and 786.0 cells (C), were cultured at a density of 750 cells/well in 384 well plates. The next day cells were cultured with increasing concentrations of monospecific DR4 antibody (pink), monospecific anti-DR5 antibody (orange), the combination of anti-DR4 and anti-DR5 antibodies (black), DR4/DR5 dual agonist Surrobody (bright green), or TRAIL (blue) for 48 hr. Relative cell viability was estimated from cellular ATP measurements using the Cell Titer Glow reagent (Promega), expressing data as % inhibition of cell survival (mean ± SEM; n= 4). In D,E and F, the experiment was performed as in A, B and C, respectfully, but the cells were pretreated with 10 nM smac mimic AEG40730 for 2h before addition of DR agonists.

Figure 3. Surrobody is a more potent inducer of caspase activation than anti-DR4 and anti-DR5 monospecific antibodies.

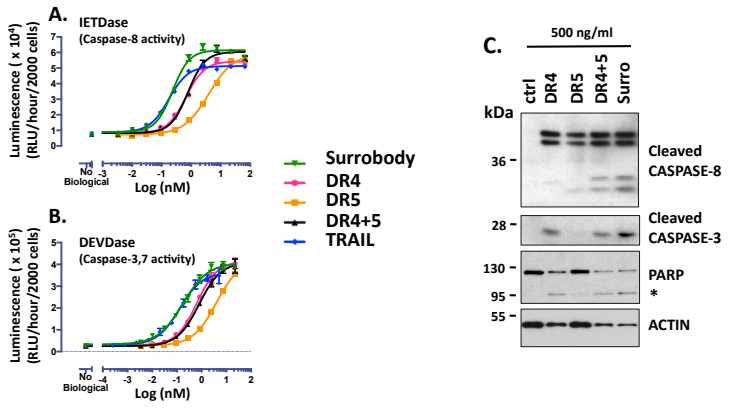
A. IETDase (Caspase-8 activity)
B. DEVDase (Caspase-3,7 activity)
Luminescence (x 10⁴) (RLU/hour/2000 cells)
Luminescence (x 10⁵) (RLU/hour/2000 cells)
No Biological
Log (nM)
Surrobody
DR4
DR5
DR4+5
TRAIL
C.
500 ng/ml
ctrl
DR4
DR5
DR4+5
Surro
kDa
36
28
130
95
55
Cleaved CASPASE-8
Cleaved CASPASE-3
PARP
*
ACTIN

MDA-MB-231 cells were cultured in 384 wells (2000 cells per well) and treated with increasing concentrations of DR agonists for 6 hr. The activity of Caspase 8 (A) and Caspase 3/7 (B) was assessed using Caspase-8 Glo (contains IETD peptide) and ApoLive Glo (contains DEVD peptide) reagents, respectively. (C) MDA-MB-231 cells were plated at a density of 70,000 cells/well and the next day they were treated for 5 h with various DR agonists at 500 ng/mL . Cells were lysed into SDS-sample buffer and lysates were analyzed by SDS-PAGE/immunoblotting using antibodies specific for cleaved Caspase-8, cleaved Caspase-3, PARP, or beta-actin.

Figure 4. Surrobody shows superior activity to anti-DR4 and anti-DR5 monospecific antibodies in 3D tumor spheroid culture model.

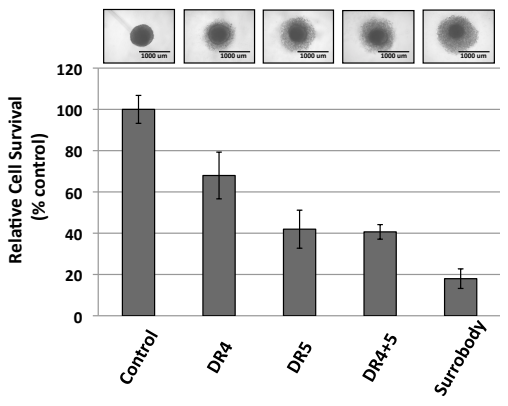
Relative Cell Survival (% control)
120
100
80
60
40
20
0
Control
DR4
DR5
DR4+5
Surrobody

MDA-MB-231 cells were cultured in ultralow attachment 96 well plates and grown for 8 days to reach a tumor spheroid diameter of ~750 um. The spheroids were then treated with indicated DR agonists at 1000 ng/ml for 2 days, then survival was assessed by measuring cellular ATP (using Cell Titer Glow reagents). Data represent mean ± SD of three spheroids. Representative phase-contrast images of spheroids are provided. Treatment with DR agonists left a core of residual surviving cells surrounded by dead and dying cells

Figure 5. DR4 and DR5 resistant tumor cell lines remain sensitive to Surrobody.

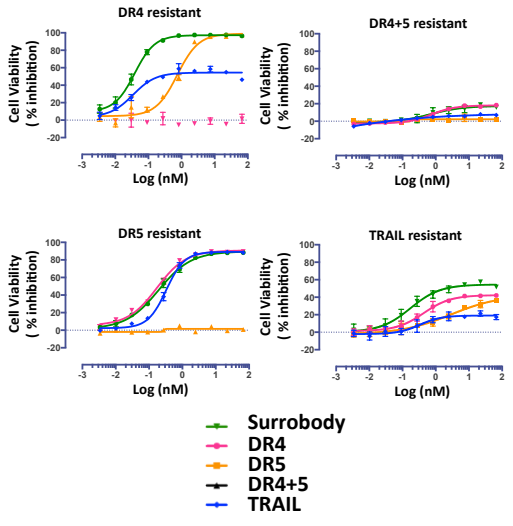
DR4 resistant
DR4+5 resistant
DR5 resistant
TRAIL resistant
Cell Viability (% inhibition)
Log (nM)
Surrobody
DR4
DR5
DR4+5
TRAIL

MDA-MB-231 cell lines resistant to DR4, DR5, DR4+DR5, or TRAIL were created by culturing them for 3 weeks in 500 ng/ml of anti-DR4 antibody, 1000ng/ml of anti-DR5 antibody, 250 ng/ml of anti-DR4 and anti-DR5, or 50 ng/ml TRAIL. To assess the sensitivity of these resistant cells to various DR agonists, the cells were plated at a density of 750 cells/well into 384 wells and cultured with increasing concentrations of anti-DR4 antibody (pink), anti-DR5 antibody (orange), DR4/DR5 dual agonist Surrobody (bright green), or TRAIL (blue) for 48 hr. Cell viability was estimated from cellular ATP measurements using the Cell Titer Glow reagent (Promega), expressing data as % inhibition of cell survival (mean ± SEM; n= 4).

Figure 6. Surrobody inhibits tumor xenograft growth in nude mice.

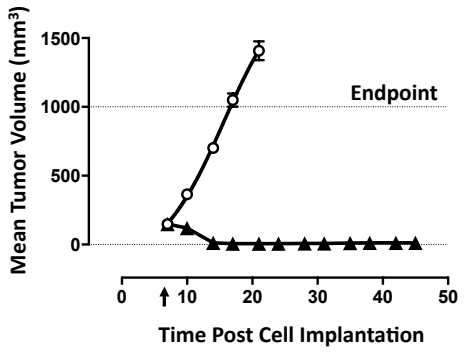
Mean Tumor Volume (mm³)
1500
1000
500
0
Endpoint
0
10
20
30
40
50
Time Post Cell Implantation

Colo-205 cells (3 x 10^6) were implanted into flanks of nude mice and were allowed to reach approximately 150 mm^3 (7 days) before starting intravenous treatment with either PBS vehicle or 3 mg/kg Surrobody. The mice were treated twice weekly between post implant day 7 and 21 and the tumor size was measured twice weekly. Mean tumor volumes and SEMs are shown for PBS treated (white circles) and Surrobody treated (filled triangles) mice (10 mice per treatment group). The tumors below palpitation limit were assigned volume of 0 mm^3. The PBS treated mice were sacrificed at day 21 when the tumors surpassed the endpoint volume of 1000 mm^3, while Surrobody treated mice were sacrificed at 45 days post-implantation.

Conclusions

We describe the generation of a novel dual specificity Surrobody that activates both death receptors DR4 and DR5. This Surrobody exhibits higher potency and efficacy than monospecific DR4 or DR5 activating antibodies in a number of cancer cell lines as well as in cells grown in 3D spheres. We also show that cell lines unresponsive to DR agonists can be sensitized to Surrobody with smac mimics such as AEG40730. Additionally, we generated DR4 and DR5 resistant cells and demonstrated that they remain sensitive to Surrobody-induced killing. Our xenograft study validated the efficacy of Surrobody in vivo and demonstrated great promise for its further development as an anti-cancer agent.

Contacts: smilutinovic@burnham.org, Ramesh.Bhatt@sealanebio.com, Michael.Horowitz@sealanebio.com

References:

1. Xu et al. PNAS (2008) 105, 10756-10761
2. Xu et al. J Mol Biol (2010) 397, 352-360

Acknowledgments:

We thank AEGERA Therapeutics for kindly providing AEG40730.